P7-053

A novel molecular mechanism for the treatment of major depressive disorder: A phase 2, proof-of-concept study of the potassium channel opener XEN1101 in subjects with moderate to severe MDD

Constanza Luzon Rosenblut, Gregory N. Beatch, Cynthia Harden, Riz Golnar, Chris Kenney

Xenon Pharmaceuticals Inc., Burnaby, BC, Canada

Background

- Major depressive disorder (MDD) is a highly prevalent mental health disorder and one of the leading causes of disability and mortality worldwide. With available treatments, 35-50% of patients fail to show a substantial improvement in symptoms, and 20-30% of patients discontinue treatment for multiple reasons, including tolerability.[1]
- The most commonly used antidepressants largely share the same mechanism of action. Therefore, a new antidepressant, acting on a different target, could potentially benefit patients who do not respond to common treatments, or who have undesirable adverse effects.

Kv7 potassium channels in depression

- Resilience is proposed to be an active coping mechanism to stress induced depression.[2] One active neural mechanism of resilience is the normalization of firing rate of ventral tegmental area (VTA) dopaminergic neurons, as observed in the chronic social defeat stress model (CSDS).[3]
- Friedman et al. demonstrated that overexpression of Kv7.3 in the VTA dopaminergic neurons normalized neuronal hyperactivity and depressive behaviors in the CSDS model in mice.[4] The same effect was achieved with local (intra-VTA) or systemic infusion of the first-generation Kv7 channel opener ezogabine.
- Enhancing the open-state of KCNQ2/3 in neurons favors a hyperpolarized resting state, which reduces rapid action potential spiking. This mechanism has been clinically proven to be effective for treatment of focal onset seizures (FOS) in adults with epilepsy with ezogabine and XEN1101.
- Both an open-label[5] and a double-blind, randomized study[6] showed significant effects of ezogabine on depression symptoms, including anhedonia, in subjects with moderate to severe MDD (Fig. 1).
- An investigator-initiated, phase 2 study of XEN1101 in MDD is ongoing at Icahn School of Medicine at Mount Sinai.

Figure 1. Results from a double-blind, placebo-controlled study of ezogabine 300 mg TID in subjects with moderate to severe MDD[6]

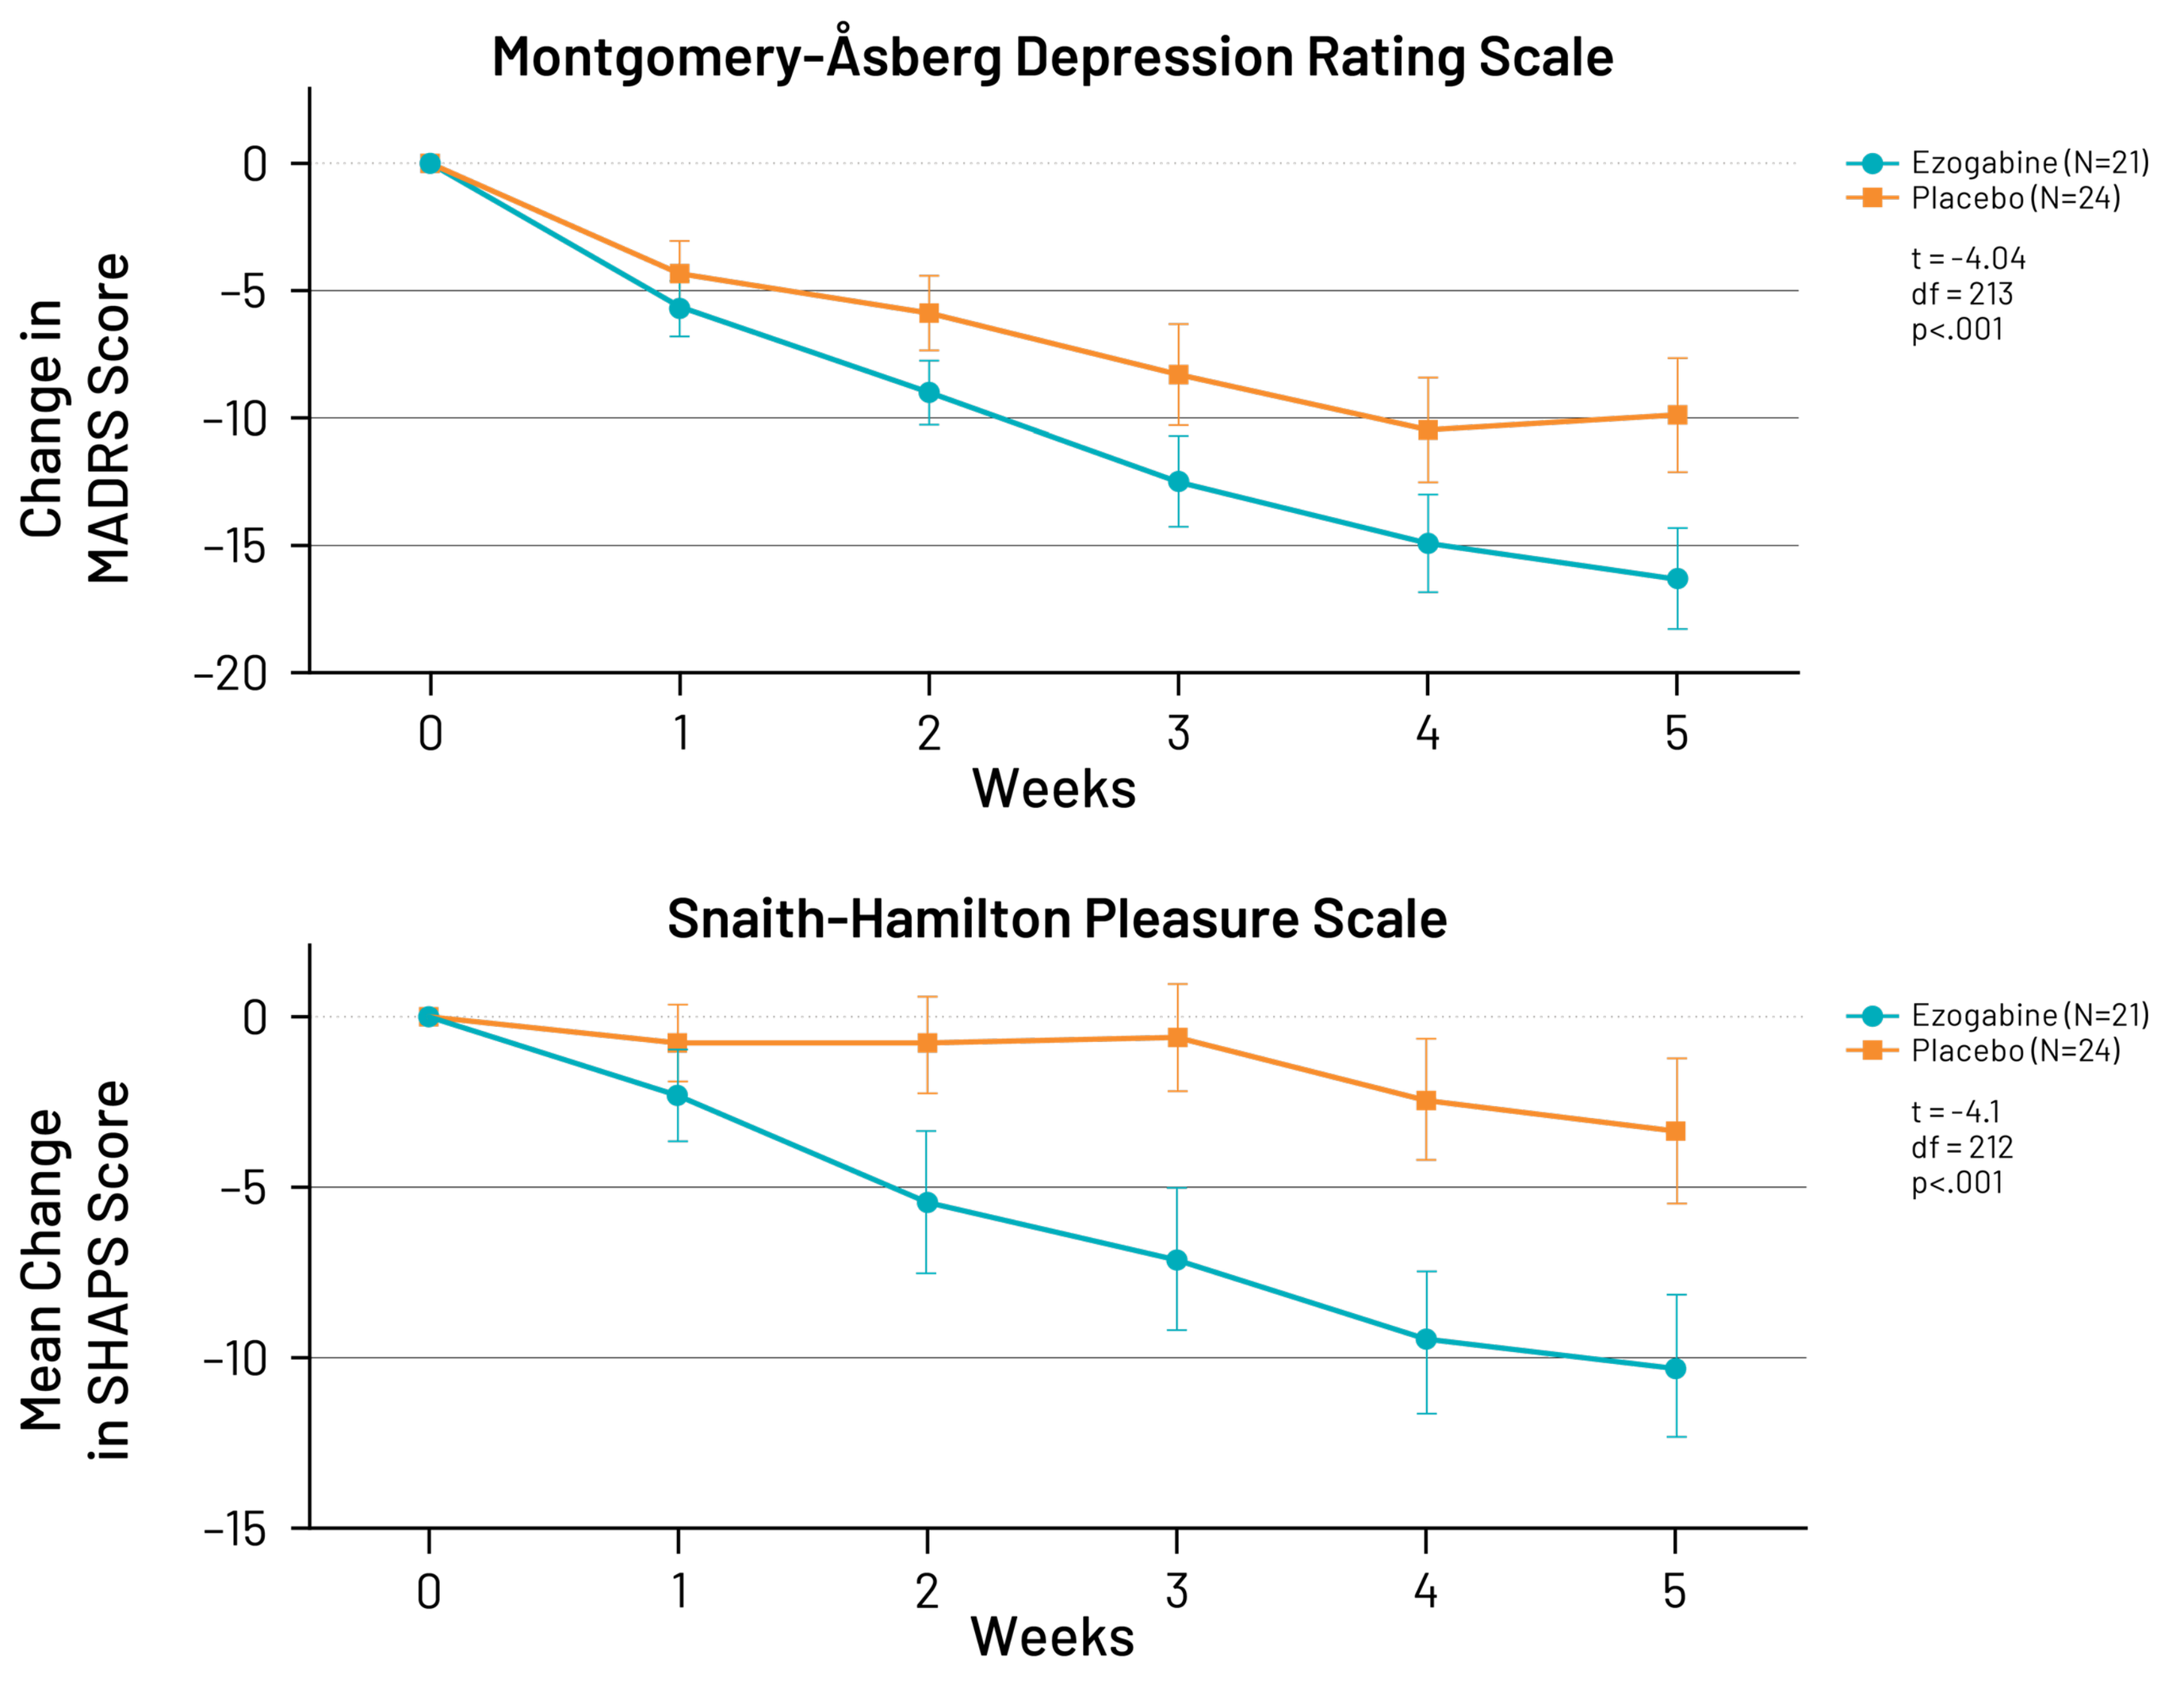

Montgomery-Åsberg Depression Rating Scale
Change in MADRS Score
0
−5
−10
−15
−20
0
1
2
3
4
5
Weeks
Ezogabine (N=21)
Placebo (N=24)
t = −4.04
df = 213
p<.001
Snaith-Hamilton Pleasure Scale
Mean Change in SHAPS Score
0
−5
−10
−15
0
1
2
3
4
5
Weeks
Ezogabine (N=21)
Placebo (N=24)
t = −4.1
df = 212
p<.001

Figures reproduced from Costi et al.[6] Depression symptoms assessed by the Montgomery-Åsberg Depression Rating Scale. Anhedonia symptoms assessed by the Snaith-Hamilton Pleasure Scale.

XEN1101

- XEN1101 is a next generation Kv7 potassium channel opener currently being developed for the treatment of epilepsy. It is formulated for once daily oral administration in the fed state.
- Depression is common in epilepsy, and it can affect quality of life and treatment adherence.[7] Moreover, some of the most commonly used antiseizure medications can cause or exacerbate mood symptoms.[8]
- Results from a recently completed large phase 2 study of XEN1101 in 325 subjects with FOS showed a significant effect in reducing seizure frequency and a favorable safety profile. It was generally well-tolerated, with dose-dependent tolerability. The most common treatment-emergent adverse events observed in this study were dizziness, somnolence, fatigue, and headache.[9]
- XEN1101 was assessed in the progressive ratio test, a protocol that evaluates motivational performance and decisional anhedonia in rats. In this test XEN1101 increased the total number of lever presses, indicating improved motivation versus vehicle (Fig. 2).

Figure 2. Results of the progressive ratio test

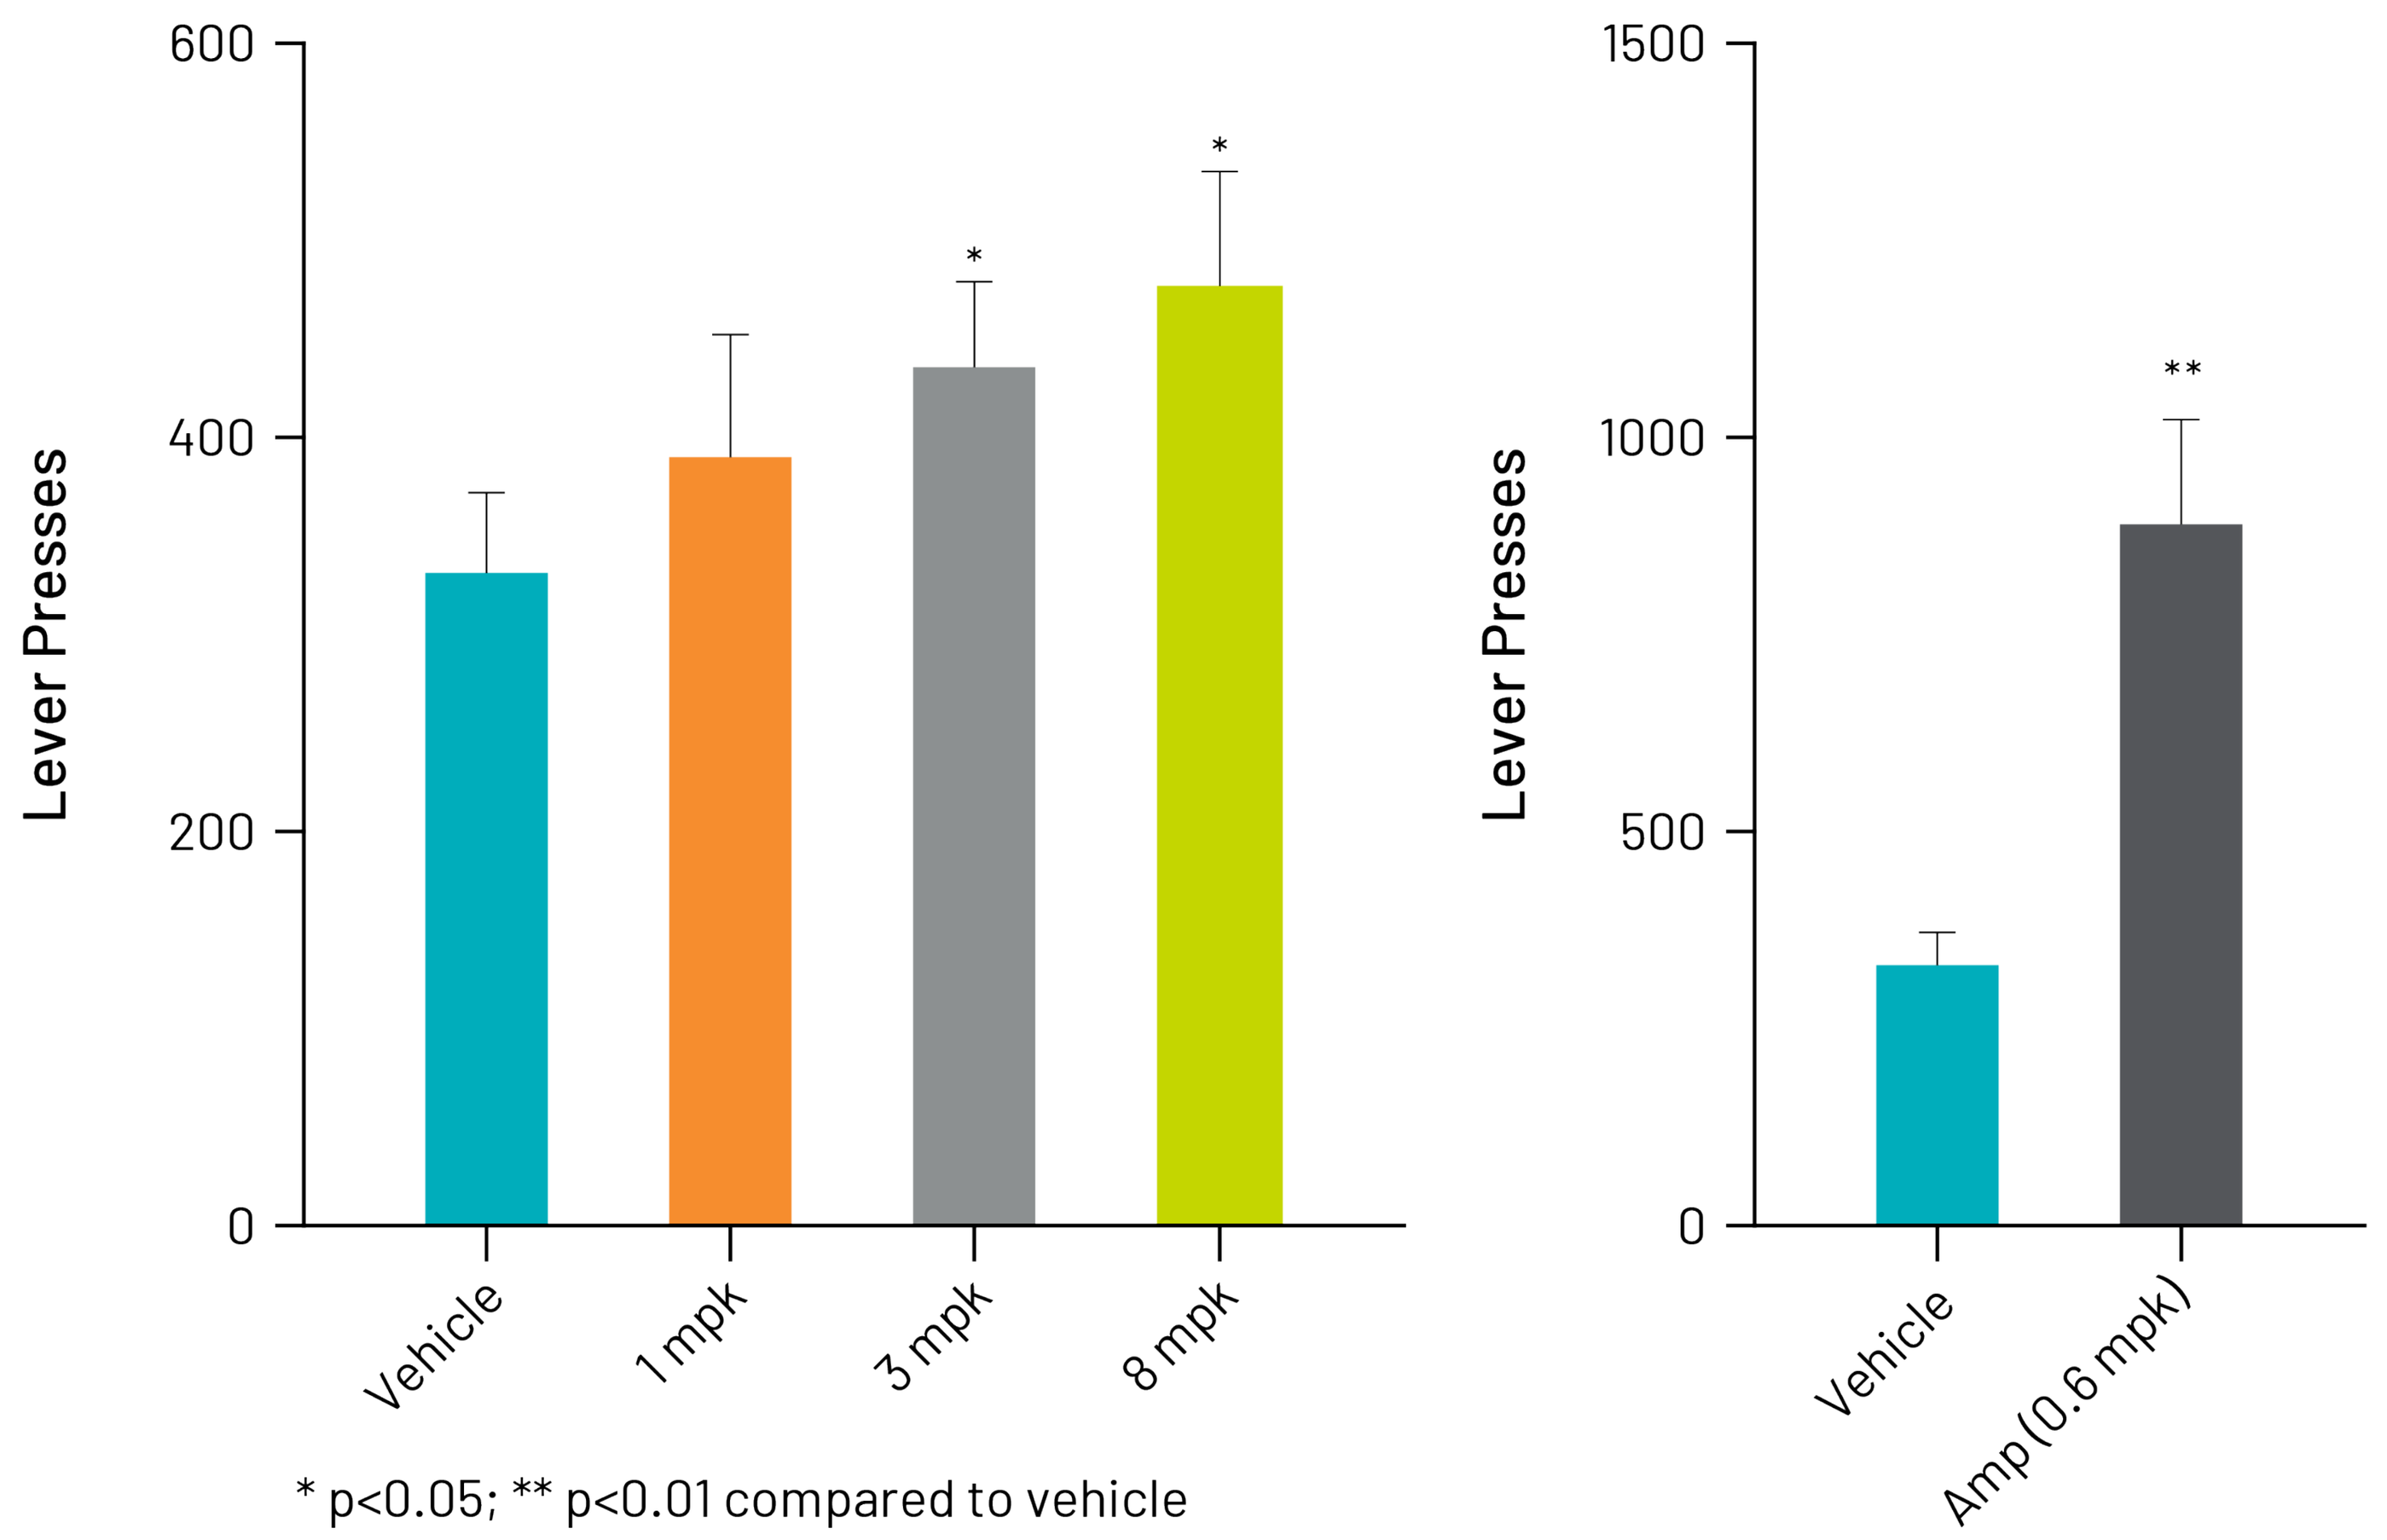

Lever Presses
600
400
200
0
Vehicle
1 mpk
3 mpk
8 mpk
*
*
Lever Presses
1500
1000
500
0
Vehicle
Amp (0.6 mpk)
**

* p<0.05; ** p<0.01 compared to vehicle

Rats were trained to respond for food made available under a progressive schedule of reinforcement in which the number of lever presses required to obtain a food reward increased for each successive reward. In a cross-over design, rats received a single dose of 1, 3, 8 mpk XEN1101 or vehicle (n=32). Total number of lever presses in the test session were measured. The stimulant amphetamine was used as a positive assay control.

- Based on the above evidence, a phase 2, double-blind, proof-of-concept study has recently started, with the objective of assessing the efficacy, safety, and tolerability of XEN1101 in subjects with moderate to severe MDD (Fig. 3).
- Approximately 150 subjects will be randomized to 3 arms: XEN1101 10 mg QD, XEN1101 20 mg QD or placebo. Dose reduction for intolerability will be allowed.
- The key eligibility criteria for the study are summarized in Table 1. Eligibility will be confirmed using the State versus Trait, Assessability, Face Validity, Ecological Validity, Rule of Three Ps (SAFER) criteria inventory performed by a remote centralized rater.[10]

Methods

Figure 3. Study design

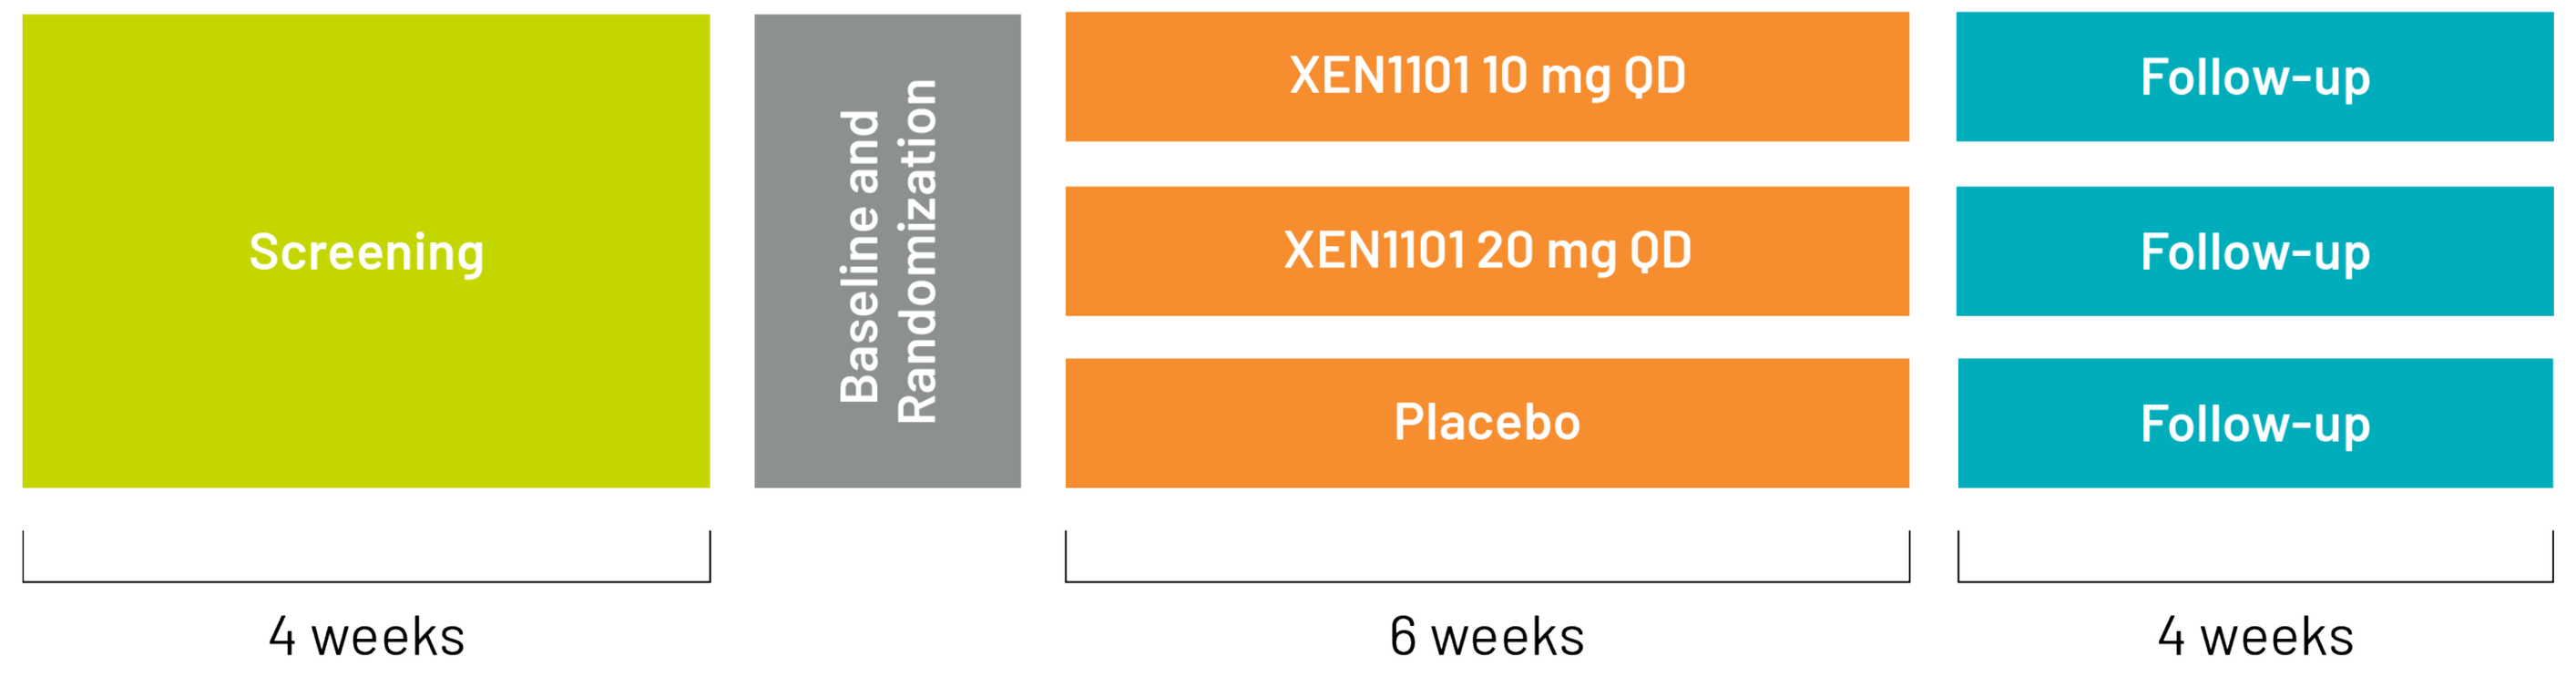

Screening
Baseline and Randomization
XEN1101 10 mg QD
XEN1101 20 mg QD
Placebo
Follow-up
Follow-up
Follow-up
4 weeks
6 weeks
4 weeks

Table 1. Key eligibility criteria

Inclusion Criteria
1. Male or female, aged 18 through 65 years (inclusive) with a body mass index (BMI) ≤35 kg/m^2.
2. Subject must meet the Diagnostic and Statistical Manual of Mental Disorders, Fifth Edition (DSM-5) criteria for major depressive disorder (MDD) and currently be experiencing a moderate to severe major depressive episode (MDE), confirmed using the Mini International Neuropsychiatric Interview (MINI).
3. Current MDE duration ≥2 months and <24 months at the time of screening.
4. Current illness severity that is at least moderate, defined as a score of ≥20 on the Hamilton Depression Rating Scale, 17-Item (HAM-D17) at screening and on Day 1.
5. Score ≥20 on the Snaith-Hamilton Pleasure Scale (SHAPS) at screening and on Day 1.
Exclusion Criteria
1. A primary psychiatric diagnosis other than MDD as defined by DSM-5 (comorbid anxiety disorders are allowed).
2. Concomitant use of antidepressants and/or other disallowed pharmacotherapy (including benzodiazepines).
3. History of schizophrenia or other psychotic disorder, MDD with psychotic features, bipolar I or II disorder, or MDD with mixed features.
4. History of non-response to >1 antidepressant drug due to lack of efficacy in the current MDE.
5. Failing >3 antidepressant drug trials, for any reason, in the current MDE.
6. Active suicidal plan/intent in the past 6 months, or >1 lifetime suicide attempt.
7. Meets criteria for a substance use disorder within the past 12 months, with the exception of tobacco use, and/or has a positive urine toxicology screen for drugs of abuse.

Study objectives and endpoints

- Primary and secondary efficacy objectives and endpoints are summarized in Table 2.
- Changes in sleep and activity parameters will be assessed using a wearable biometric device. Subjects will wear the device on their non-dominant wrist from randomization to the last follow up visit, 24 hours a day.
- Safety assessments include adverse events and serious adverse events, clinical laboratory tests, ECGs, vital signs, Columbia–Suicide Severity Rating Scale, and the Clinical Institute Withdrawal Assessment Scale–Benzodiazepines to evaluate withdrawal symptoms after the end of treatment. Dilated ophthalmological exams and the American Urological Association Symptom Index will also be conducted to assess for retinal pigment epithelium abnormalities and symptoms of urinary retention, respectively, as these effects were observed during treatment of epilepsy patients with ezogabine.
- The plasma levels of XEN1101 will also be evaluated.

Table 2. Efficacy objectives and endpoints

Objectives	Endpoints
Primary	
• To assess the efficacy of 10 mg and 20 mg doses of XEN1101 compared to placebo on improvement of depressive symptoms.	• Montgomery-Åsberg Depression Rating Scale (MADRS) score change through Week 6.
Secondary	
• To assess the efficacy of XEN1101 compared to placebo on improvement of anhedonia symptoms.	• Snaith-Hamilton Pleasure Scale (SHAPS) score change through Week 6.
• To assess the efficacy of XEN1101 compared to placebo on improvement of anxiety symptoms.	• Beck Anxiety Inventory (BAI) score change through Week 6.
Exploratory	
• To assess the effect of XEN1101 compared to placebo on depressive symptoms.	• Quick Inventory of Depressive Symptomatology, Self-Report (QIDS-SR) score change through Week 6. • Responder rate at Week 6, defined as the percentage of subjects with ≥50% reduction in MADRS score. • Percentage of subjects in remission (MADRS ≤10) at Week 6. • Hamilton Depression Rating Scale, 17-Item (HAM-D17) score change through Week 6. • MADRS score, SHAPS score, BAI score, QIDS-SR score, and HAM-D17 score at all timepoints and change from baseline to all timepoints.
• To assess the effect of XEN1101 compared to placebo on overall health status.	• Clinical Global Impression of Improvement (CGI-I) score at Week 6. • Clinical Global Impression of Severity (CGI-S) score change at Week 6. • Patient Global Impression of Improvement (PGI-I) score at Week 6.

CONCLUSIONS

- XEN1101 represents a novel molecular mechanism to potentially treat depression. The double-blind, placebo controlled, proof-of-concept study described above will help evaluate the efficacy and safety of this compound in subjects with MDD and inform further development.

References

1. Cipriani A, Furukawa TA, Salanti G, et al. Comparative efficacy and acceptability of 12 new-generation antidepressants: a multiple-treatments meta-analysis. *Lancet*. 2009;373(9665):746-58.
2. Russo SJ, Murrough JW, Han M, Charney DS, Nestler EJ. Neurobiology of resilience. *Nat Neurosci*. 2012; 15(11): 1475–1484. doi: 10.1038/nn.3234.
3. Krishnan V, et al. Molecular adaptations underlying susceptibility and resistance to social defeat in brain reward regions. *Cell*. 2007; 131:391–404. doi: 10.1016/j.cell.2007.09.018.
4. Friedman AK, Juarez B, Ku SM et al. KCNQ channel openers reverse depressive symptoms via an active resilience mechanism. *Nat Commun*. 2016; 7(11671). doi: 10.1038/ncomms11671.
5. Tan A, Costi S, Morris LS, et al. Effects of the KCNQ channel opener ezogabine on functional connectivity of the ventral striatum and clinical symptoms in patients with major depressive disorder. *Mol Psychiatry*. 2020; 25:1323–1333. doi: 10.1038/s41380-018-0283-2.
6. Costi S, Morris LS, Kirkwood KA et al. Impact of the KCNQ2/3 channel opener ezogabine on reward circuit activity and clinical symptoms in depression: results from a randomized controlled trial. *Am J Psychiatry*. 2021; 178(5):437-446. doi: 10.1176/appi.ajp.2020.20050653.
7. Kanner AM, Schachter SC, Barry JJ et al. Depression and epilepsy: epidemiologic and neurobiologic perspectives that may explain their high comorbid occurrence. *Epilepsy Behav*. 2012; 24(2): 156-68. doi: 10.1016/j.yebeh.2012.01.007.
8. Mula M, Sander JW. Negative effects of antiepileptic drugs on mood in patients with epilepsy. *Drug Saf*. 2007; 30(7): 555-67. doi: 10.2165/00002018-200730070-00001.
9. French J, Porter R et al. Phase 2b efficacy and safety of XEN1101, a novel potassium channel modulator, in adults with focal epilepsy (X-TOLE). Poster presented at: 2021 Annual Meeting of the American Epilepsy Society. 2021 Dec 3-7; Chicago, IL.
10. Desseilles M, Witte J, Chang TE et al. Massachusetts General Hospital SAFER criteria for clinical trials and research. *Harv Rev Psychiatry*. 2013; 21(5): 269-74. doi: 10.1097/HRP.0b013e3182a75cc7.

Supported by funding from Xenon Pharmaceuticals Inc. The presented information concerns a use that has not been approved by the US Food and Drug Administration.

XENON